BioPharma Product Testing

Accelerating Advanced Therapy Medicinal Product (ATMP) Release with Platform Bioassays: Smarter Strategies for Impurity Control on AAV and LVV products.

By Davide Medici, Luca Benedan, Arianna Spagliardi, Simone Gallinea, Giulia Mancini

Abstract

Robust impurity control is essential in viral vector manufacturing to ensure patient safety, process consistency, and timely release of ATMPs. Across AAV and LVV production processes, recurring impurities such as residual endonuclease and host cell proteins require analytical methods capable of delivering consistent, reliable performance under routine manufacturing conditions.

This work presents a platform analytical strategy developed and validated in accordance with ICH guidelines, with specific focus on how the validation approach enables broad applicability across multiple viral vector products. The platform concept was implemented by harmonising method design, predefined acceptance criteria, and system suitability requirements, allowing a single validation exercise to support multiple matrices and manufacturing platforms.

Two rapid platform methods were developed: a quantitative residual endonuclease assay and a host cell protein (HCP) assay, with rapid turnaround times. The validation program included cross matrix evaluation, robustness studies reflecting routine operational variability, and performance assessments focused on accuracy, repeatability, precision, and linearity. This ensured that each method met predefined platform acceptance criteria and remained suitable for both in process monitoring and QC release testing. Additionally , both assays were designed to operate with minimal sample consumption, an important advantage in gene therapy testing where material availability is often limited and must be conserved for critical development and release activities.

By integrating validated platform methods into viral vector workflows, manufacturers can streamline method lifecycle management, enhance impurity trending, and accelerate ATMP release.

ATMPs overview

ATMPs are classified into three main groups according to Reg (EC) No. 1394/2007:
- Gene therapy Medicinal products (*in vivo* or *ex vivo)*
- Somatic Cell Therapy Products
- Tissue Engineered Products

ATMPs can be tailored-made for the individual patient (autologous: cells directly collected from the patients, treated and re-introduced into the patient itself) or manufactured for larger population (allogenic: cells from healthy donor, treated, expanded, and introduced into multiple end patients)

Like all medicines, ATMP products both for clinical trials and for commercial intent must align with the regulation related to Good Manufacturing Practices (GMP).

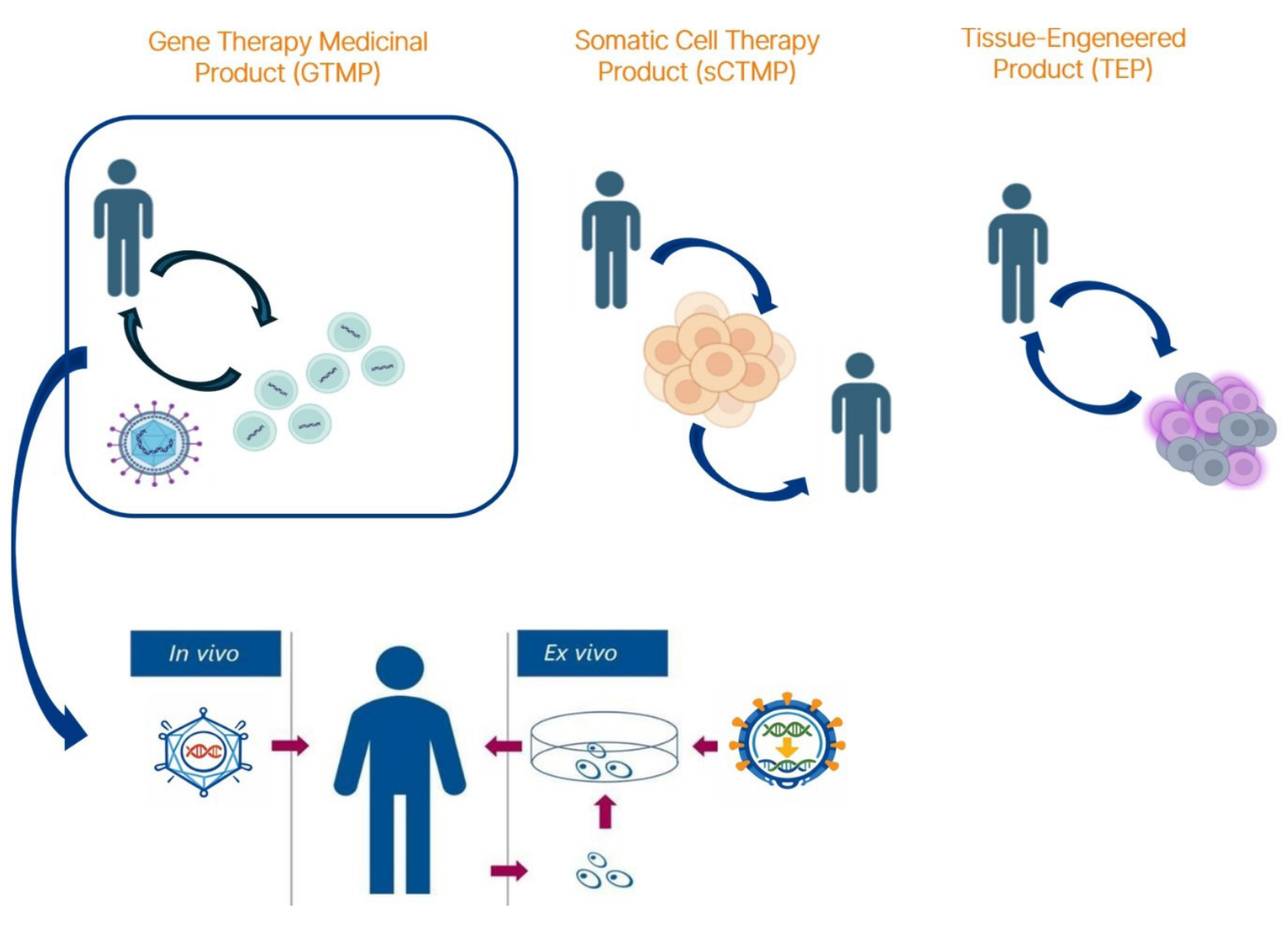

Gene Therapy Medicinal Product (GTMP)
Somatic Cell Therapy Product (sCTMP)
Tissue-Engeneered Product (TEP)
In vivo
Ex vivo

ATMPs quality attributes

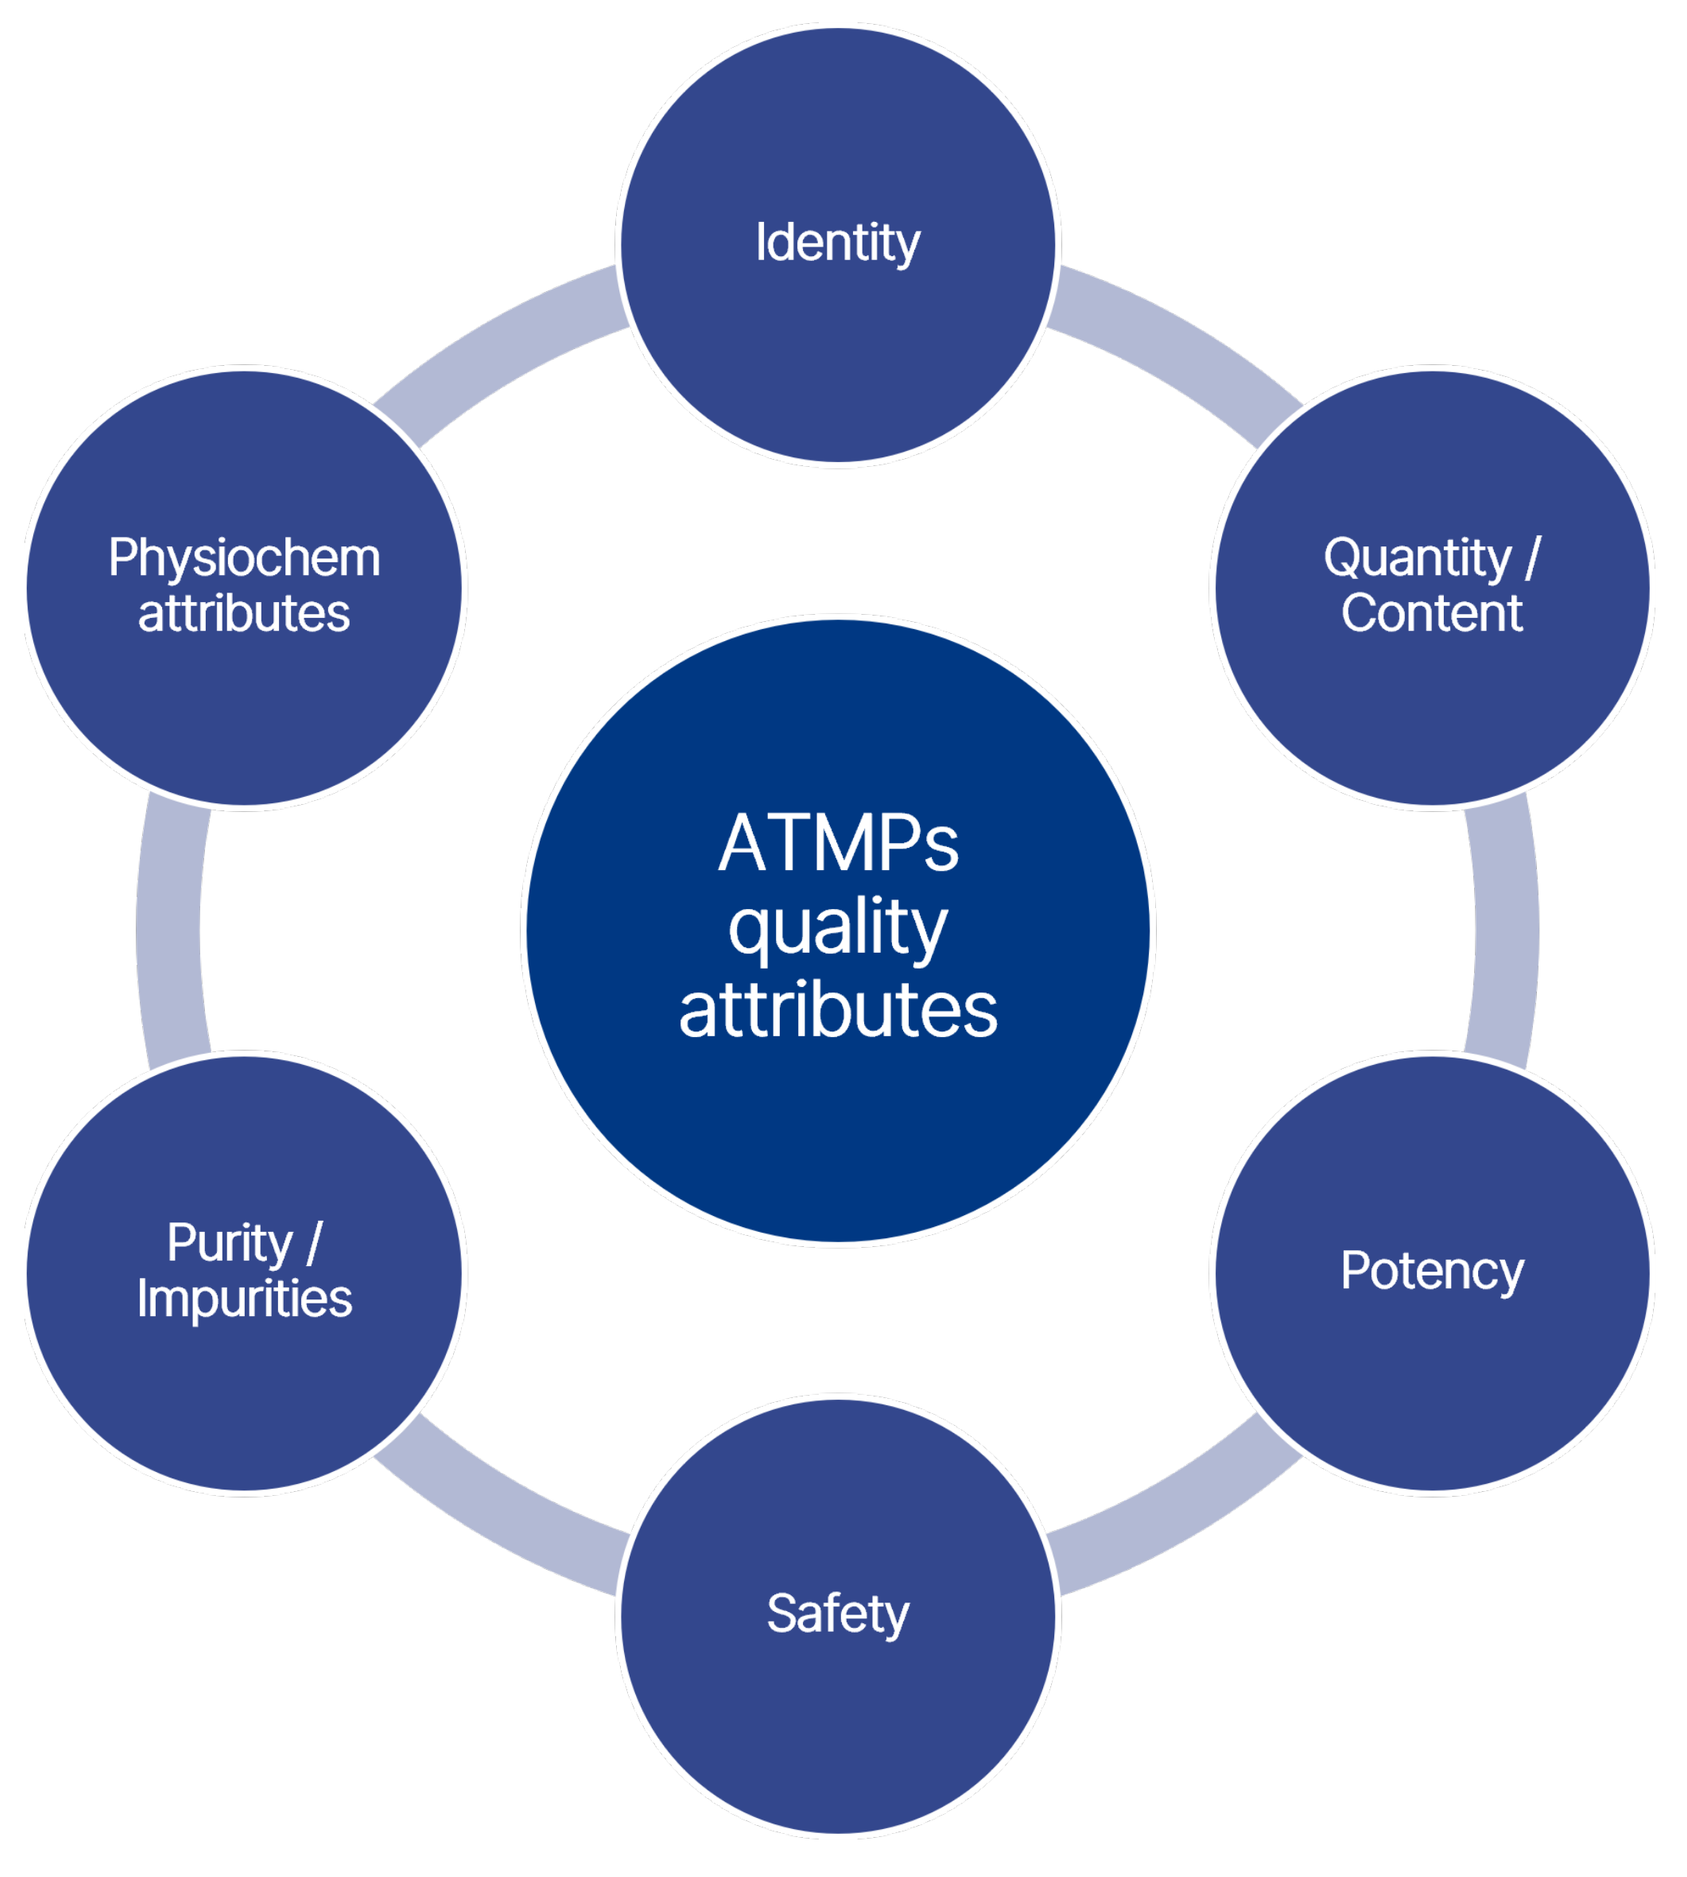

ATMPs quality attributes
Identity
Quantity / Content
Potency
Safety
Purity / Impurities
Physiochem attributes

ATMPs challenges for testing

1. Product complexity
 - Highly complex and heterogeneous products
 - Batch to batch variability, especially autologus products
2. Limited sample avaiability
 - Small batch sizes
 - Limited materials for QC release
3. Turnaround time constrains
 - Short shelf life of some ATMPs
 - Rapid release testing requiered
4. Analytical method development
 - Lack of standardised or compendial methods
 - Product specific and process specific assays
 - Assays often complex, low throughput and time-consuming
5. Method validation challenges
 - Limited reference materials
 - Difficulty applying classical validation approaches
6. Impurity control
 - Product related impurities
 - Process related Impurities (HCP and Residual Endonuclaese)

Among these challenges, impurity control emerges as a key critical aspect, due to the intrinsic complexity of ATMPs and the presence of multiple product- and process-related contaminants. This becomes even more relevant in viral vector manufacturing, where impurities such as host cell proteins, residual DNA, and enzymes must be accurately dentified and quantified using reliable analytical methods.

Viral vectors production produce different impurities

During the manufacturing of an investigational ATMP, varying levels of product- and process-related impurities may be introduced into the active substance. Regulatory authorities require that such impurities—ranging from process contaminants (e.g., media components, growth factors, host cell proteins and DNA, residual enyzmes) to product-related species (e.g., unintended cell types, non-viable cells, fragments, precursors, or degradation products)—be minimised from the earliest development stages. Applicants must provide a risk assessment and demonstrate the use of appropriate analytical methods to identify, monitor, and control these impurities.

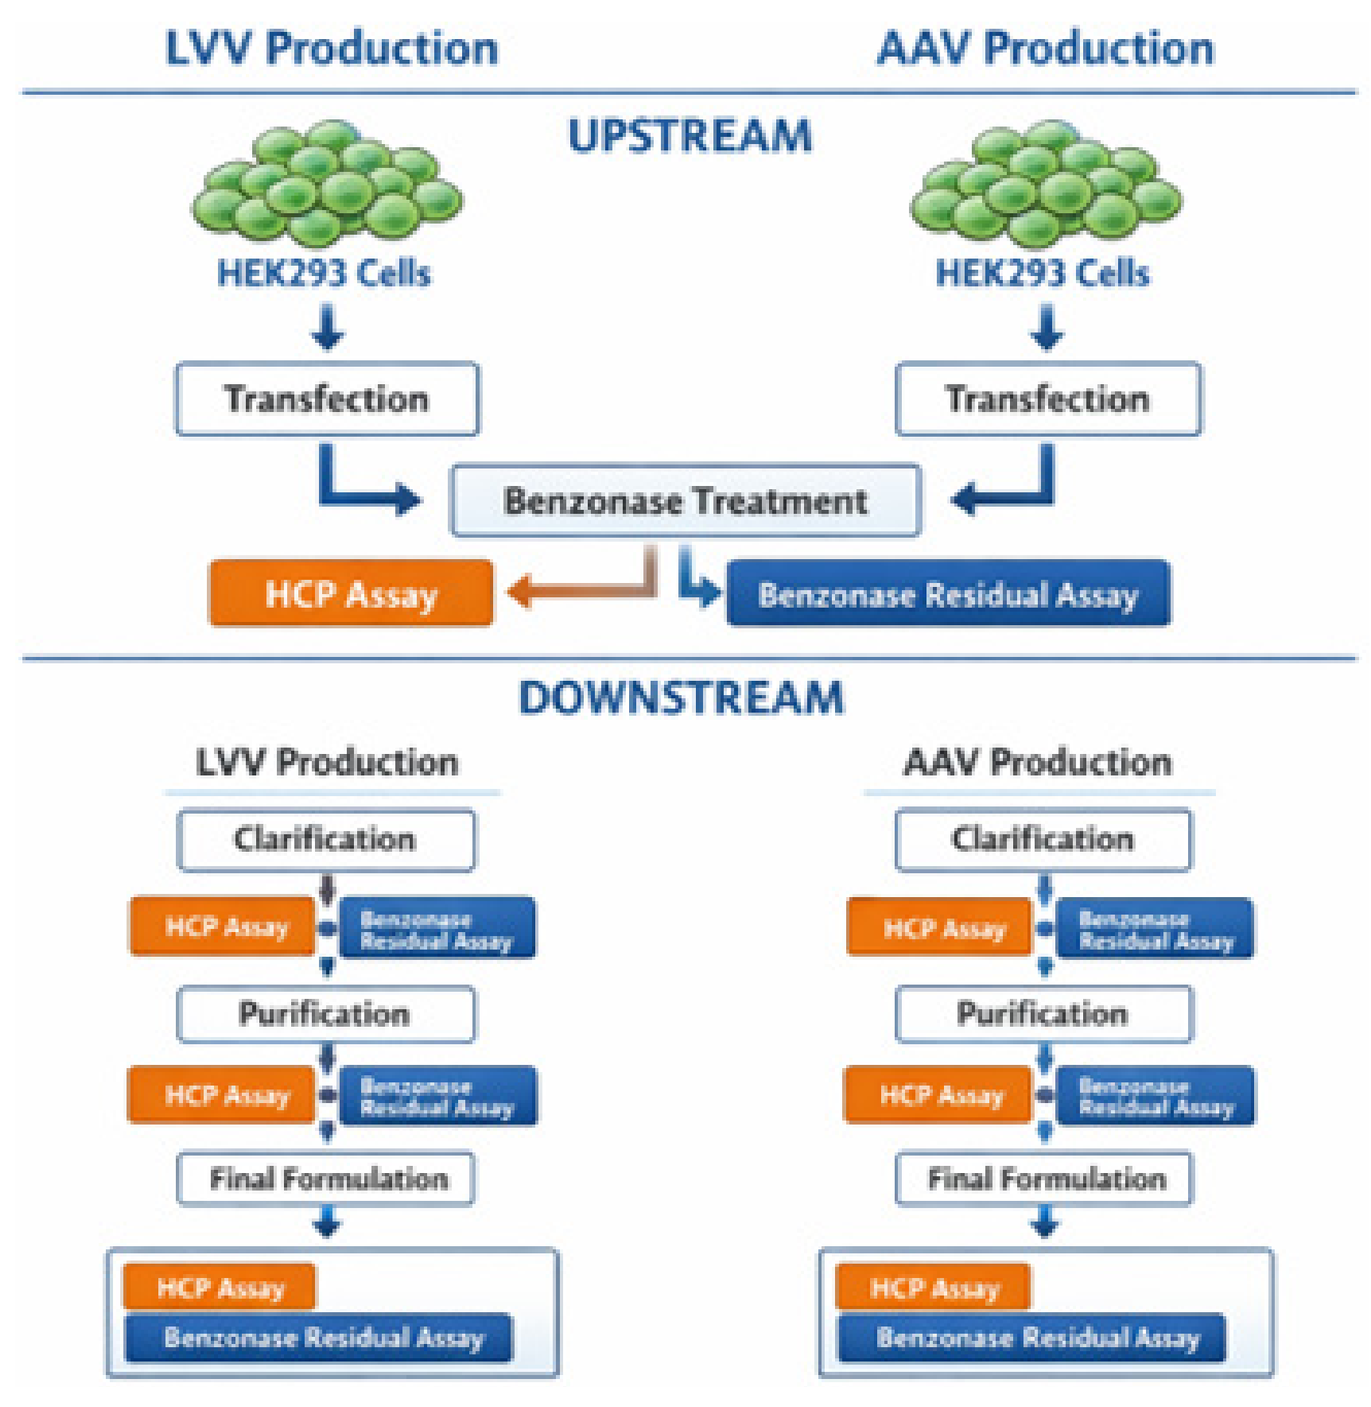

LVV Production
AAV Production
UPSTREAM
HEK293 Cells
HEK293 Cells
Transfection
Transfection
Benzonase Treatment
HCP Assay
Benzonase Residual Assay
DOWNSTREAM
LVV Production
AAV Production
Clarification
HCP Assay
Benzonase Residual Assay
Purification
Final Formulation

These challenges require innovative QC testing strategies to ensure product availability and patient safety

Analytical Platform method

Process & Product related impurities on LVV & AAV vectors production

Product-Related Impurities: Impurities that are structurally related to the intended product and are generated during production, purification, or storage, but differ from the desired product in identity, quality, or purity.

Process-Related Impurities: Impurities that are introduced during the manufacturing process and are derived from process inputs such as host cells, raw materials, reagents, or equipment, and are not intended components of the final product.

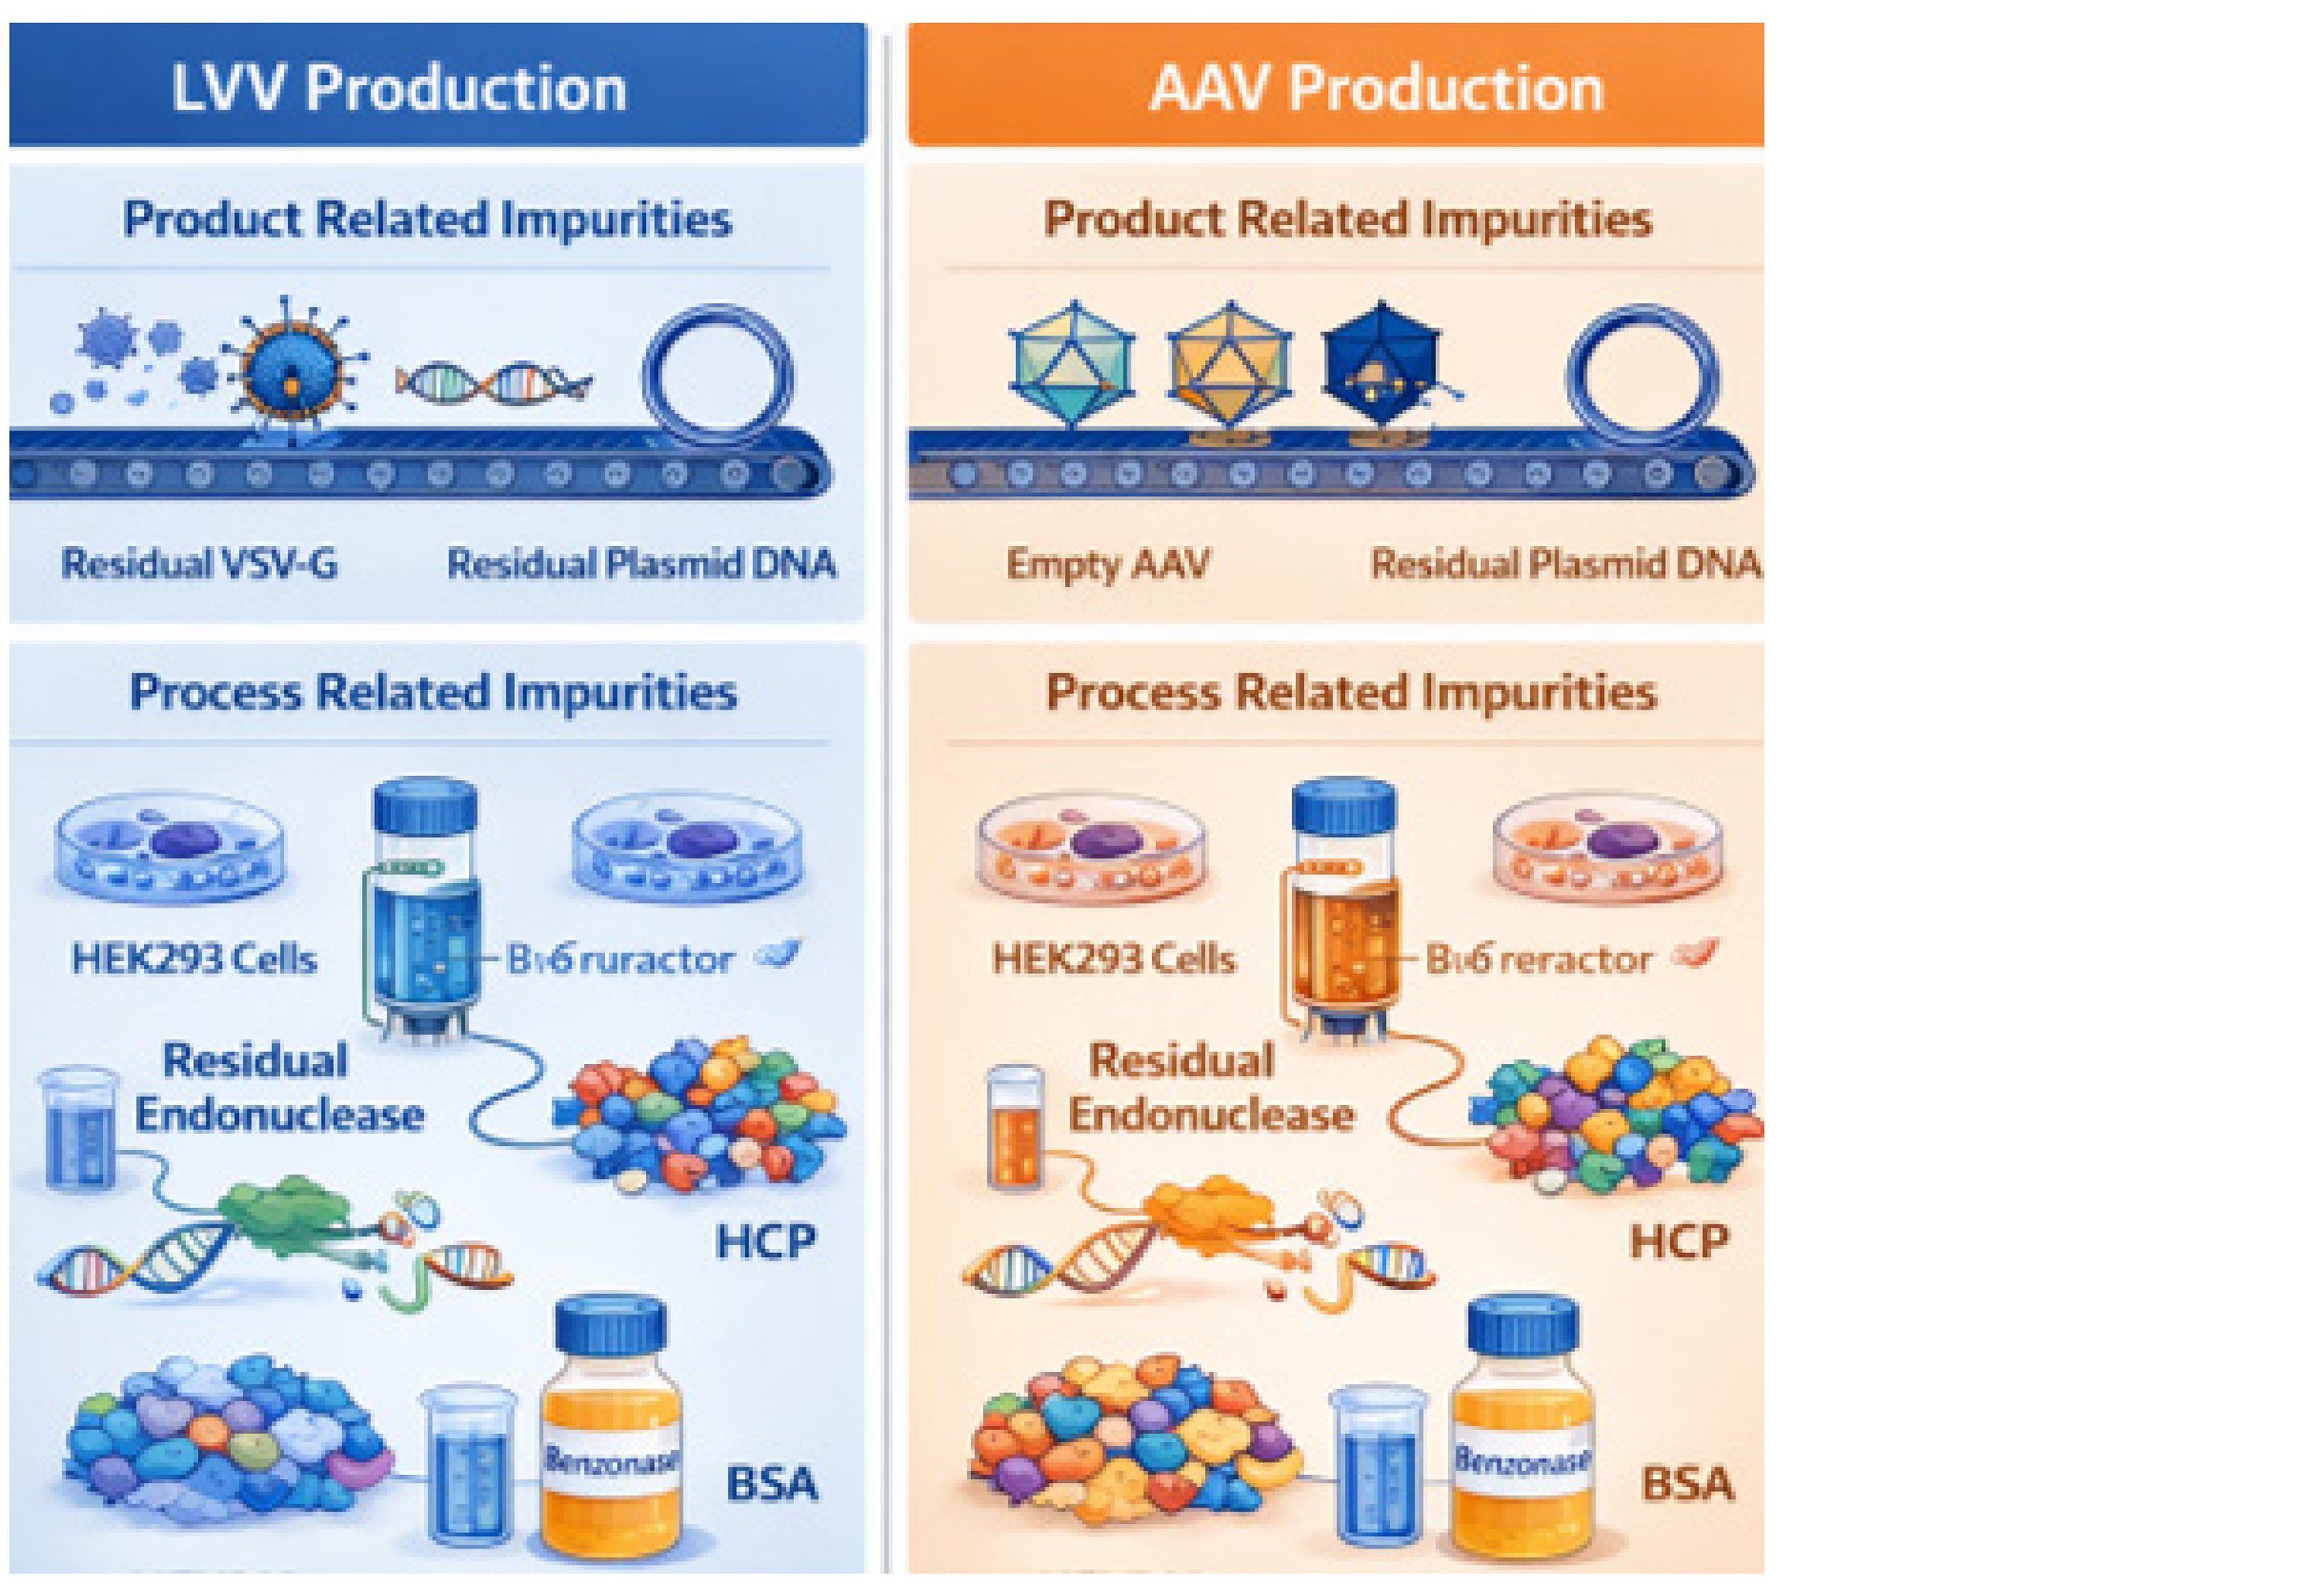

LVV Production
AAV Production
Product Related Impurities
Residual VSV-G
Residual Plasmid DNA
Empty AAV
Residual Plasmid DNA
Process Related Impurities
HEK293 Cells
Residual Endonuclease
HCP
BSA
Benzonase

What is a platform?

A platform is an analytical procedure that is suitable to be applied to different products that share predefined similarities without significant change to its operational conditions.

The use of platform can reduce development and validation efforts to be used for multiple products, reducing time and material requirements.

Eurofins platform methods are provided as plug-and-play solutions, with suitability assessments performed to tailor the analytical method to the client's sample.

The platform-based method enables reduced sample consumption during client- and product-specific method development and validation, thereby addressing and optimizing one of the key challenges in the ATMP field.

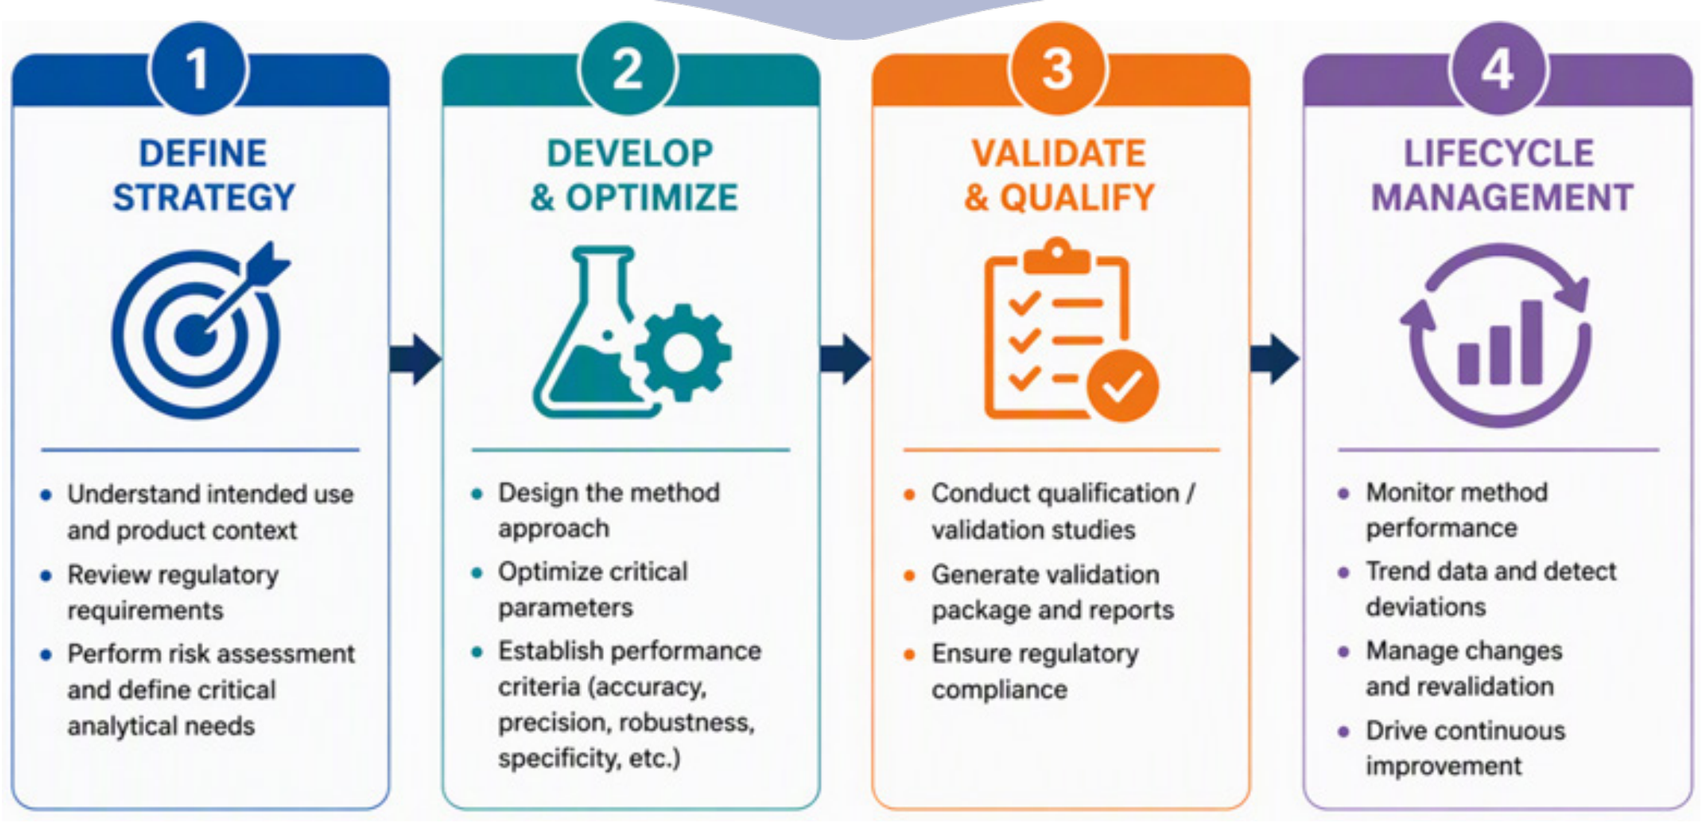

1 DEFINE STRATEGY
• Understand intended use and product context
• Review regulatory requirements
• Perform risk assessment and define critical analytical needs
2 DEVELOP & OPTIMIZE
• Design the method approach
• Optimize critical parameters
• Establish performance criteria (accuracy, precision, robustness, specificity, etc.)
3 VALIDATE & QUALIFY
• Conduct qualification / validation studies
• Generate validation package and reports
• Ensure regulatory compliance
4 LIFECYCLE MANAGEMENT
• Monitor method performance
• Trend data and detect deviations
• Manage changes and revalidation
• Drive continuous improvement

Which EBPT stakeholders participate to the analytical platform method development?

- Analytical team — Create & optimise method proposed by analytical team
- Analytical team — Review regolatory compliance and acceptance criteria — Regulatory consultants
- Analytical team — Define platform method qualification/validation studies — Regulatory consultants
- Analytical team — Execute studies and generate documentation by analytical team
- Analytical team ↔ Regulatory consultants
- Ensure regulatory documents are defendable

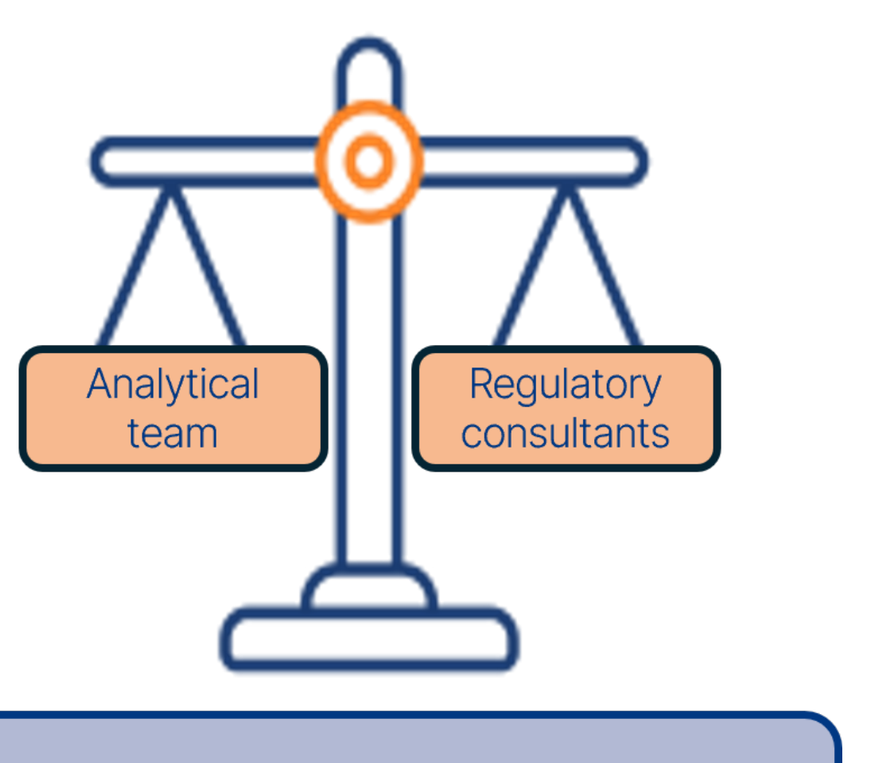

Analytical team
Regulatory consultants

Platform method development must be a synergic work between different team

Parameters evaluated and Validation

Target of quantification: HEK293 Host Cell Proteins and Total Residual Benzonase

ICH Q2 (R2) Parameter	Set up strategy
Specificity	Evaluated using data from all analytical runs by comparing the lowest standard level with the blank.
Accuracy	5 independent runs at 3 spike levels (High, Medium, Low).
Linearity	3 independent runs, minimum 5 serially diluted points, R^2 evaluation.
Repeatability	3 independent K+ ELISA preparations, same plate, same analytical session, same analyst.
Intermediate Precision	6 independent K+ ELISA preparations, different plates, different analytical sessions, ≥ 2 analysts involved.
LOD	Determined using a signal-to-noise approach.
LOQ	Evaluated in at least 3 independent runs.
Robustness	• Incubation time variation • Plate reading at different times after stop solution addition • Manual vs automated plate washer

Qualification method results strategy

Parameter	Platform Method	Performance Results
Accuracy	Host Cell Proteins	Method is accurate
	Residual Benzonase	Method is accurate
Limit of Detection (LOD)	Host Cell Proteins	S/N ≥ 2.0
	Residual Benzonase	S/N ≥ 2.0
Specificity	Host Cell Proteins	Method is specific
	Residual Benzonase	Method is specific
Repeatablity (Intra assay)	Host Cell Proteins	CV% ≤3%
	Residual Benzonase	CV% ≤10%
Intermediate Precision (Inter assay)	Host Cell Proteins	CV% ≤6%
	Residual Benzonase	CV% ≤14%
Linearity	Host Cell Proteins	R^2 = 1.00
	Residual Benzonase	R^2 = 1.00
Limit of Quantification (LOQ)	Host Cell Proteins	LOQ = 12 ng/ml
	Residual Benzonase	LOQ = 0.4 U/ml
Robustness	Host Cell Proteins	• Incubation time: Method is robust • Different timing of reading: Method is not robust • Washing modality: Method is robust
	Residual Benzonase	• Incubation time: Method is not robust • Different timing of reading: Method is robust • Washing modality: Method is robust

EBPT approach to assess analytical platform suitability

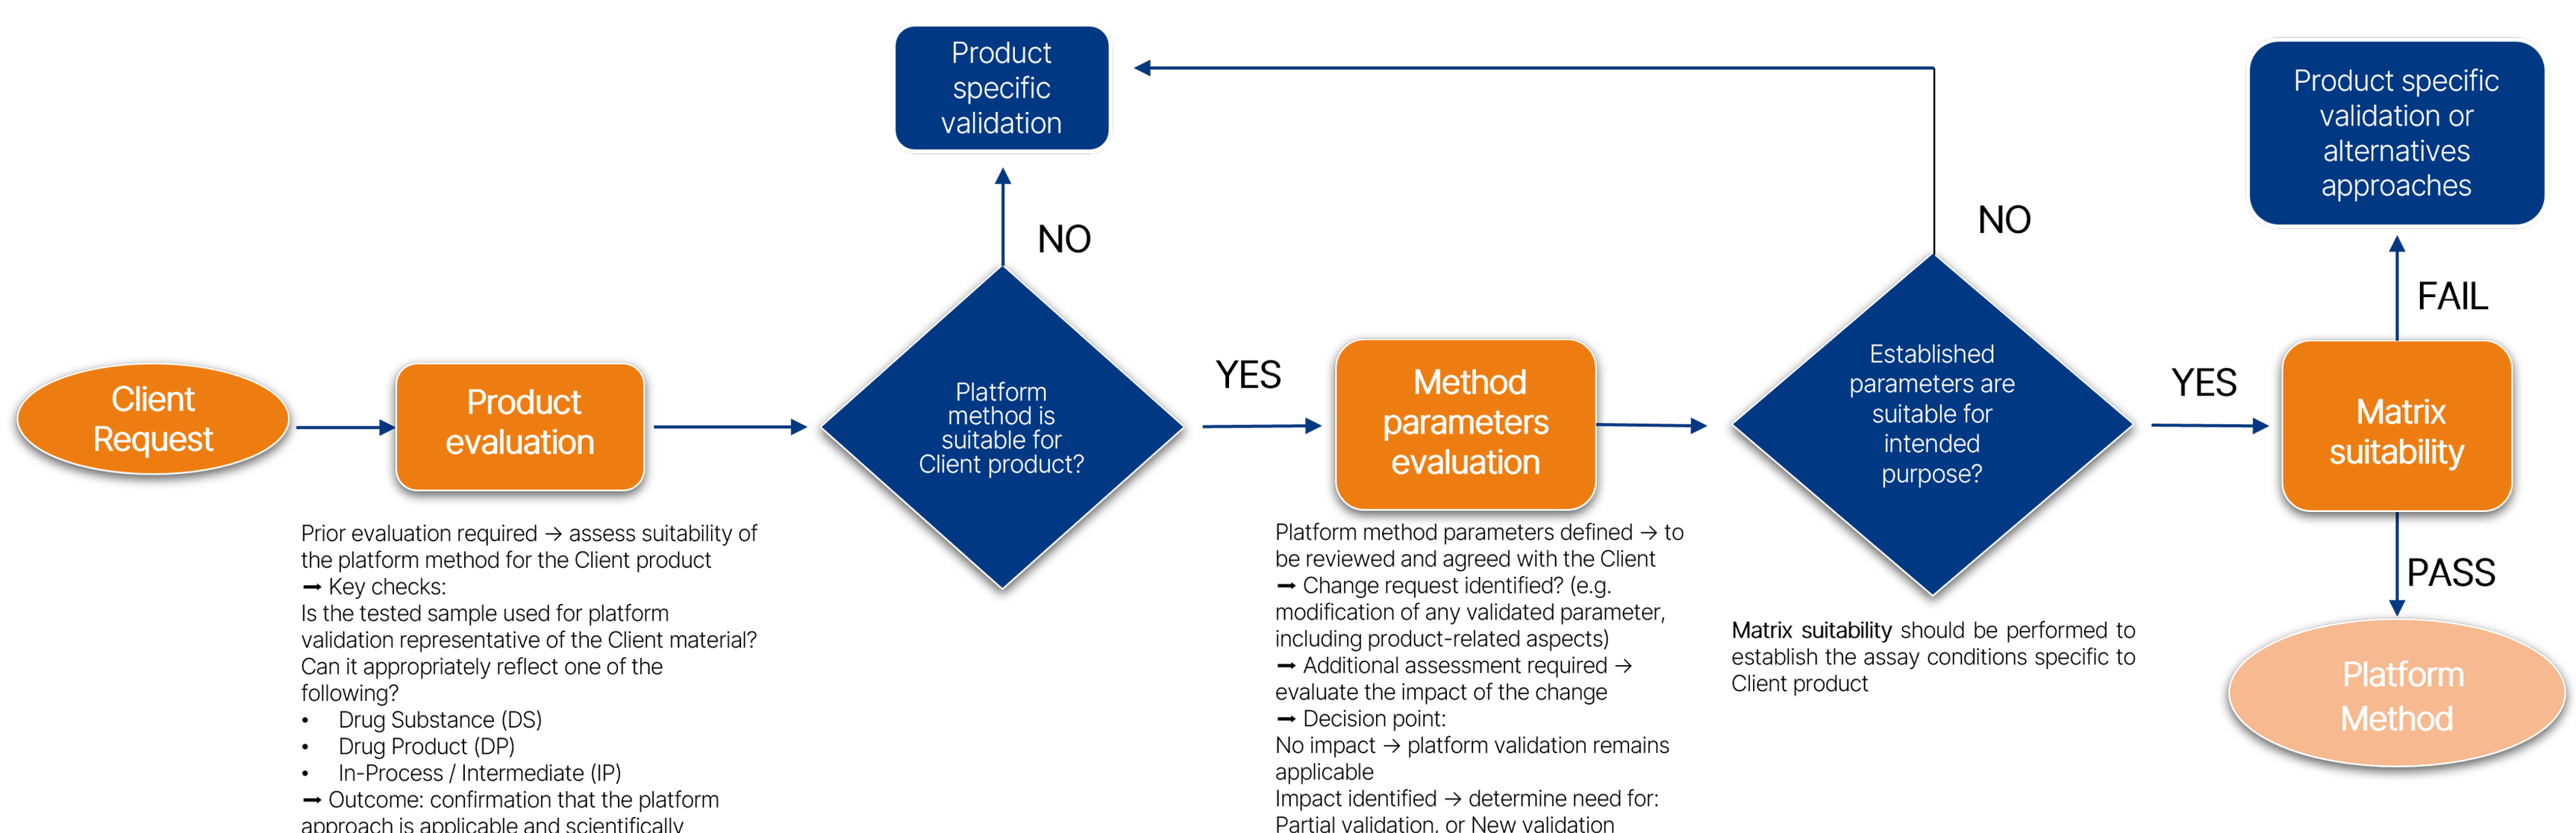

Client Request
Product evaluation
Platform method is suitable for Client product?
NO
Product specific validation
YES
Method parameters evaluation
Established parameters are suitable for intended purpose?
NO
YES
Matrix suitability
FAIL
Product specific validation or alternatives approaches
PASS
Platform Method

Prior evaluation required → assess suitability of the platform method for the Client product
➞ Key checks:
Is the tested sample used for platform validation representative of the Client material?
Can it appropriately reflect one of the following?
- Drug Substance (DS)
- Drug Product (DP)
- In-Process / Intermediate (IP)

➞ Outcome: confirmation that the platform approach is applicable and scientifically justified for the proposed Client process.

Platform method parameters defined → to be reviewed and agreed with the Client
➞ Change request identified? (e.g. modification of any validated parameter, including product-related aspects)
➞ Additional assessment required → evaluate the impact of the change
➞ Decision point:
No impact → platform validation remains applicable
Impact identified → determine need for: Partial validation, or New validation

Matrix suitability should be performed to establish the assay conditions specific to Client product

Strategy to assess the applicability - suitability of ELISA platform method on client's sample

Parameter	What it assessed	How to perform	Typical acceptance criteria	Decision notes
Matrix Interference Test	Effect of buffer/ excipients	Compare client buffer vs control (spiked)	Recovery	Critical for detergents (e.g. Tween, Pluronic)
Spike Recovery (Spiking study)	Matrix effect / accuracy in client sample	Spike standard (HCP or Benzonase) into sample at 3 levels (low/mid/high)	Recovery	Critical parameter: if it fails → method not suitable or higher MRD required
Dilution Linearity	Matrix effect and proportionality	Serial dilutions of the sample (e.g. 1:2, 1:4, 1:8...) → at least 6 serial points	Recovery and CV	Non-linearity indicates interference → method adaptation required
Precision (intra-run)	Repeatability within a run	Replicates of the same sample	CV	Required for formal suitability claim
Precision (inter-run)	Reproducibility between runs	Compare results across independent runs (different days/operators)	CV	Required for formal suitability claim
MRD (Minimum Required Dilution)	Minimum dilution to remove matrix interference	Identify dilution where recovery is acceptable	Recovery within criteria	Key output of suitability assessment

- ✓ Minimum of three independent analytical runs
- ✓ Preferably three aliquots per batch

End-to-End Analytical Platform Expertise Supporting Bioproduction

Platform Method	Adenovirus Early Region 1A (E1A)	Simian Virus 40 (SV40)	Residual Plasmid DNA (Kanamycin)	Residual Plasmid DNA (Kanamycin)	LVV Genome Copy (GC) Titer	Replication Competent LV (RCL)	HEK293 Host cell Protein	Total residual Benzonase
Quantitative or Qualitative	Quantitative	Quantitative	Quantitative	Quantitative	Quantitative	Qualitative	Quantitative	Quantitative
Scope of the test	Impurity test	Impurity test	Impurity test	Potency/ Safety test	Potency test	Safety test	Impurity test	Impurity test
Readout	qPCR	qPCR	qPCR	ddPCR	ddPCR	qPCR	ELISA assay	ELISA assay
Applicability on	AAV - LVV	AAV - LVV	AAV - LVV	LVV	LVV	LVV	AAV - LVV	AAV - LVV

- Adenovirus E1A/SV40,
- Residual plasmid DNA (Kanamycin),
- LVV GC titer,
- HEK293 host cell protein

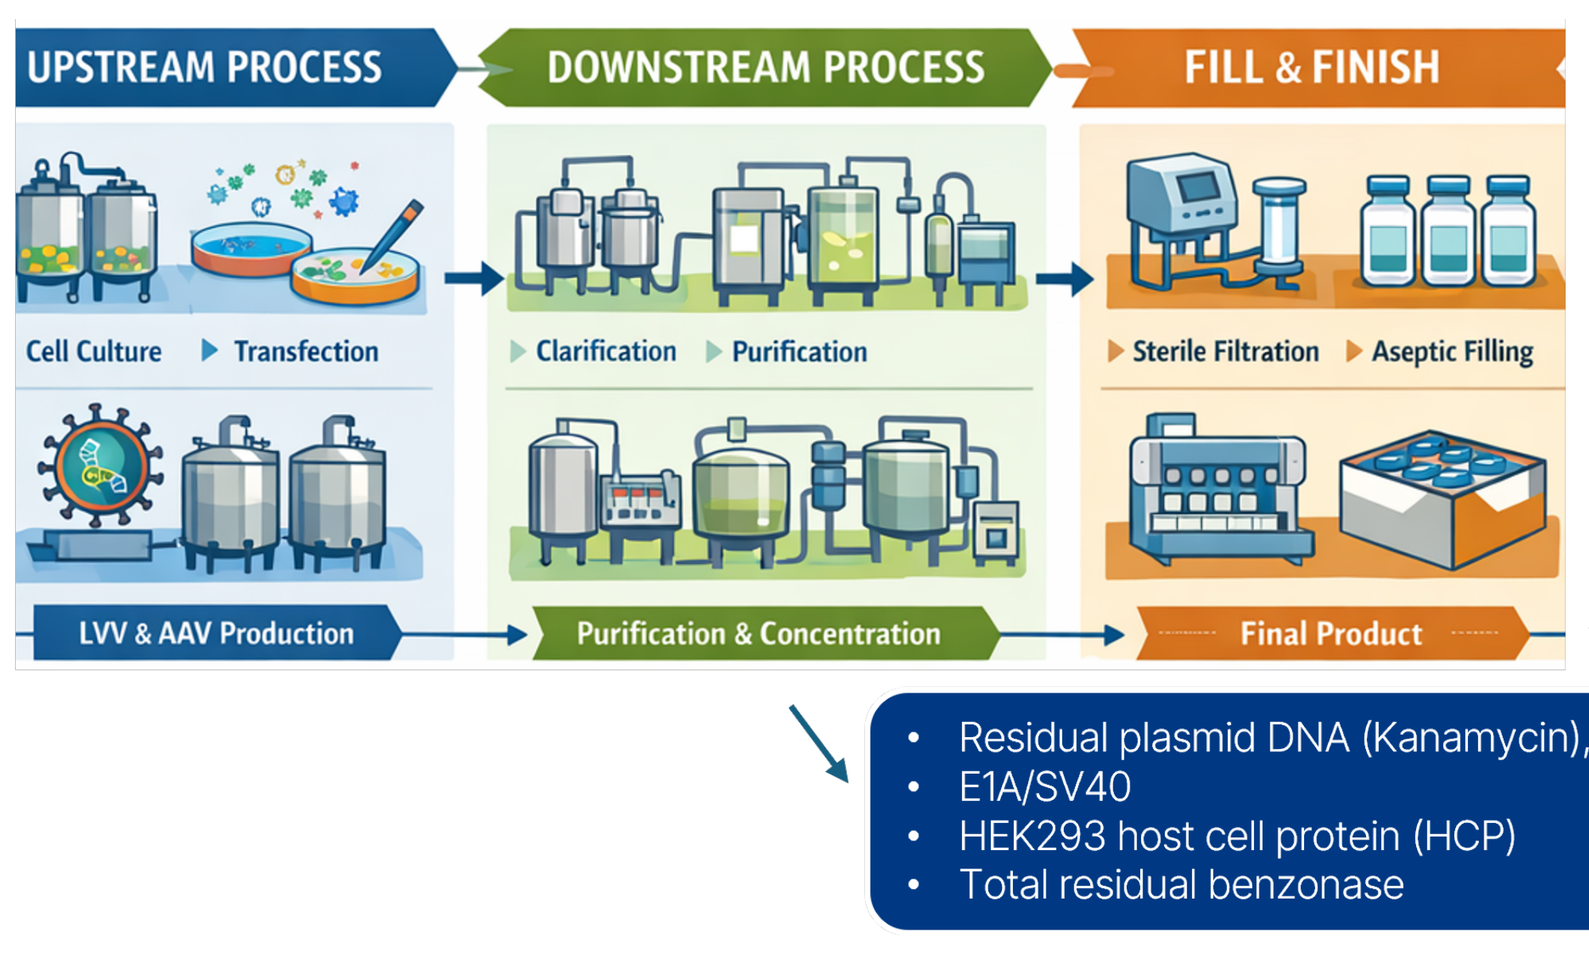

UPSTREAM PROCESS
DOWNSTREAM PROCESS
FILL & FINISH
Cell Culture
Transfection
Clarification
Purification
Sterile Filtration
Aseptic Filling
LVV & AAV Production
Purification & Concentration
Final Product

- Residual plasmid DNA (Kanamycin),
- E1A/SV40
- HEK293 host cell protein (HCP)
- Total residual benzonase

- Adenovirus E1A/SV40,
- Residual plasmid DNA (Kanamycin),
- LVV GC titer,
- LVV Vector Copy Number (VCN)
- Replication Competent LV (RCL)
- HEK293 host cell protein
- Total residual benzonase

Conclusion and take-home message

ATMPs are promising therapies but due to their complexity and challenges, innovative testing procedures and approaches are strongly needed to support their sustainability and affordability.

Current analytical testing methods are most product-specific related which contribute to increase associated timelines, costs and sample requirements.

Eurofins BPT Italy has successfully developed and qualified a range of platform methods for the characterisation and release of LVV and AAV vectors. These methods encompass key testing areas, including impurities, potency, and safety. The established platform approaches enhance the availability of standardised analytical methods and Eurofins BPT Italy has developed and validated different analytical platform methods to quantify process/product residual impurities, VCN and GC Titer promoting the avaiability of analytical methods and reducing the time needed for developing analytical procedures from scratch, significantly reduce the time required to develop new analytical procedures from scratch.

A method suitability assessment will be performed on product-specific sample to confirm the applicability of the platform method, with a tailored evaluation based on the specific matrix and intended use.

Eurofins BioPharma Product Testing Italy - Via Bruno Buozzi, 2, Vimodrone (MI)
EurofinsBiolab@BPT.eurofinseu.com
www.eurofins.com/it-it/bpt